Safety, efficacy, and PKPD of 23ME-00610, a first-in-class anti-CD200R1 antibody, in patients with advanced or metastatic ovarian cancer: results from a multi-center multi-country Phase 1/2a expansion cohort.

Ali Raza KHAKI[1], Albiruni Ryan Abdul RAZAK[2], Scott A. LAURIE[3], Ching-Chang HWANG[4], Anh N. DIEP[4], Maike SCHMIDT[4], Roo VOLD[4], Daniel MASLYAR[4], Dylan M. GLATT[4], Julie KRYSTAL[5]

[1]Stanford University, Stanford, CA, USA; [2]Princess Margaret Cancer Centre, University of Toronto, Toronto, CA, USA; [3]Ottawa Hospital Cancer Centre, University of Ottawa, Ottawa, ON, Canada; [4]23andMe, South San Francisco, CA, USA; [5]Cohen Children's Medical Center, Queens, NY, USA

#5575

POSTER PRESENTED AT ASCO 2024

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

BACKGROUND

- CD200R1 was identified as a promising immuno-oncology (IO) target from the 23andMe database [1]. Pleiotropic causal variants with opposing effect on risks for cancer and immune diseases, referred to as an IO signature, were observed for 3 critical components of the CD200R1 pathway, including CD200R1, its sole ligand CD200, and the downstream signaling protein DOK2.
- CD200R1 is expressed on immune cells and binds to CD200, its only known ligand in humans. Binding CD200 by CD200R1 can downregulate proinflammatory cytokines by activated T and myeloid cells and promote an immunosuppressive microenvironment in human cancers, where CD200 is highly expressed [2-9] (**Figure 1**).
- 23ME-00610 ('610) is a first-in-class IgG1 antibody that binds CD200R1 with high affinity and inhibits immunosuppressive signaling, leading to restoration of T cell activity and killing of CD200-expressing tumor cells in preclinical studies [1] (**Figure 1**).
- 23ME-00610 is currently in the Phase 2a portion of a Phase 1/2a clinical trial in participants with advanced solid malignancies (NCT05199272) (**Figure 2**). From the 28 patient Phase 1 portion, 23ME-00610 had acceptable safety and tolerability, a favorable PK profile supporting Q3W dosing, full target engagement with peripheral saturation at doses ≥ 60 mg, and pharmacodynamic evidence of activity, including on-target immune-related AEs, › 50% stable disease rate, and tumor shrinkage [10-12].

Figure 1. 23ME-00610, a Fully Humanized, Effectorless IgG1, Inhibits Immunosuppressive CD200/R1 Signaling via High Affinity Binding to CD200R1

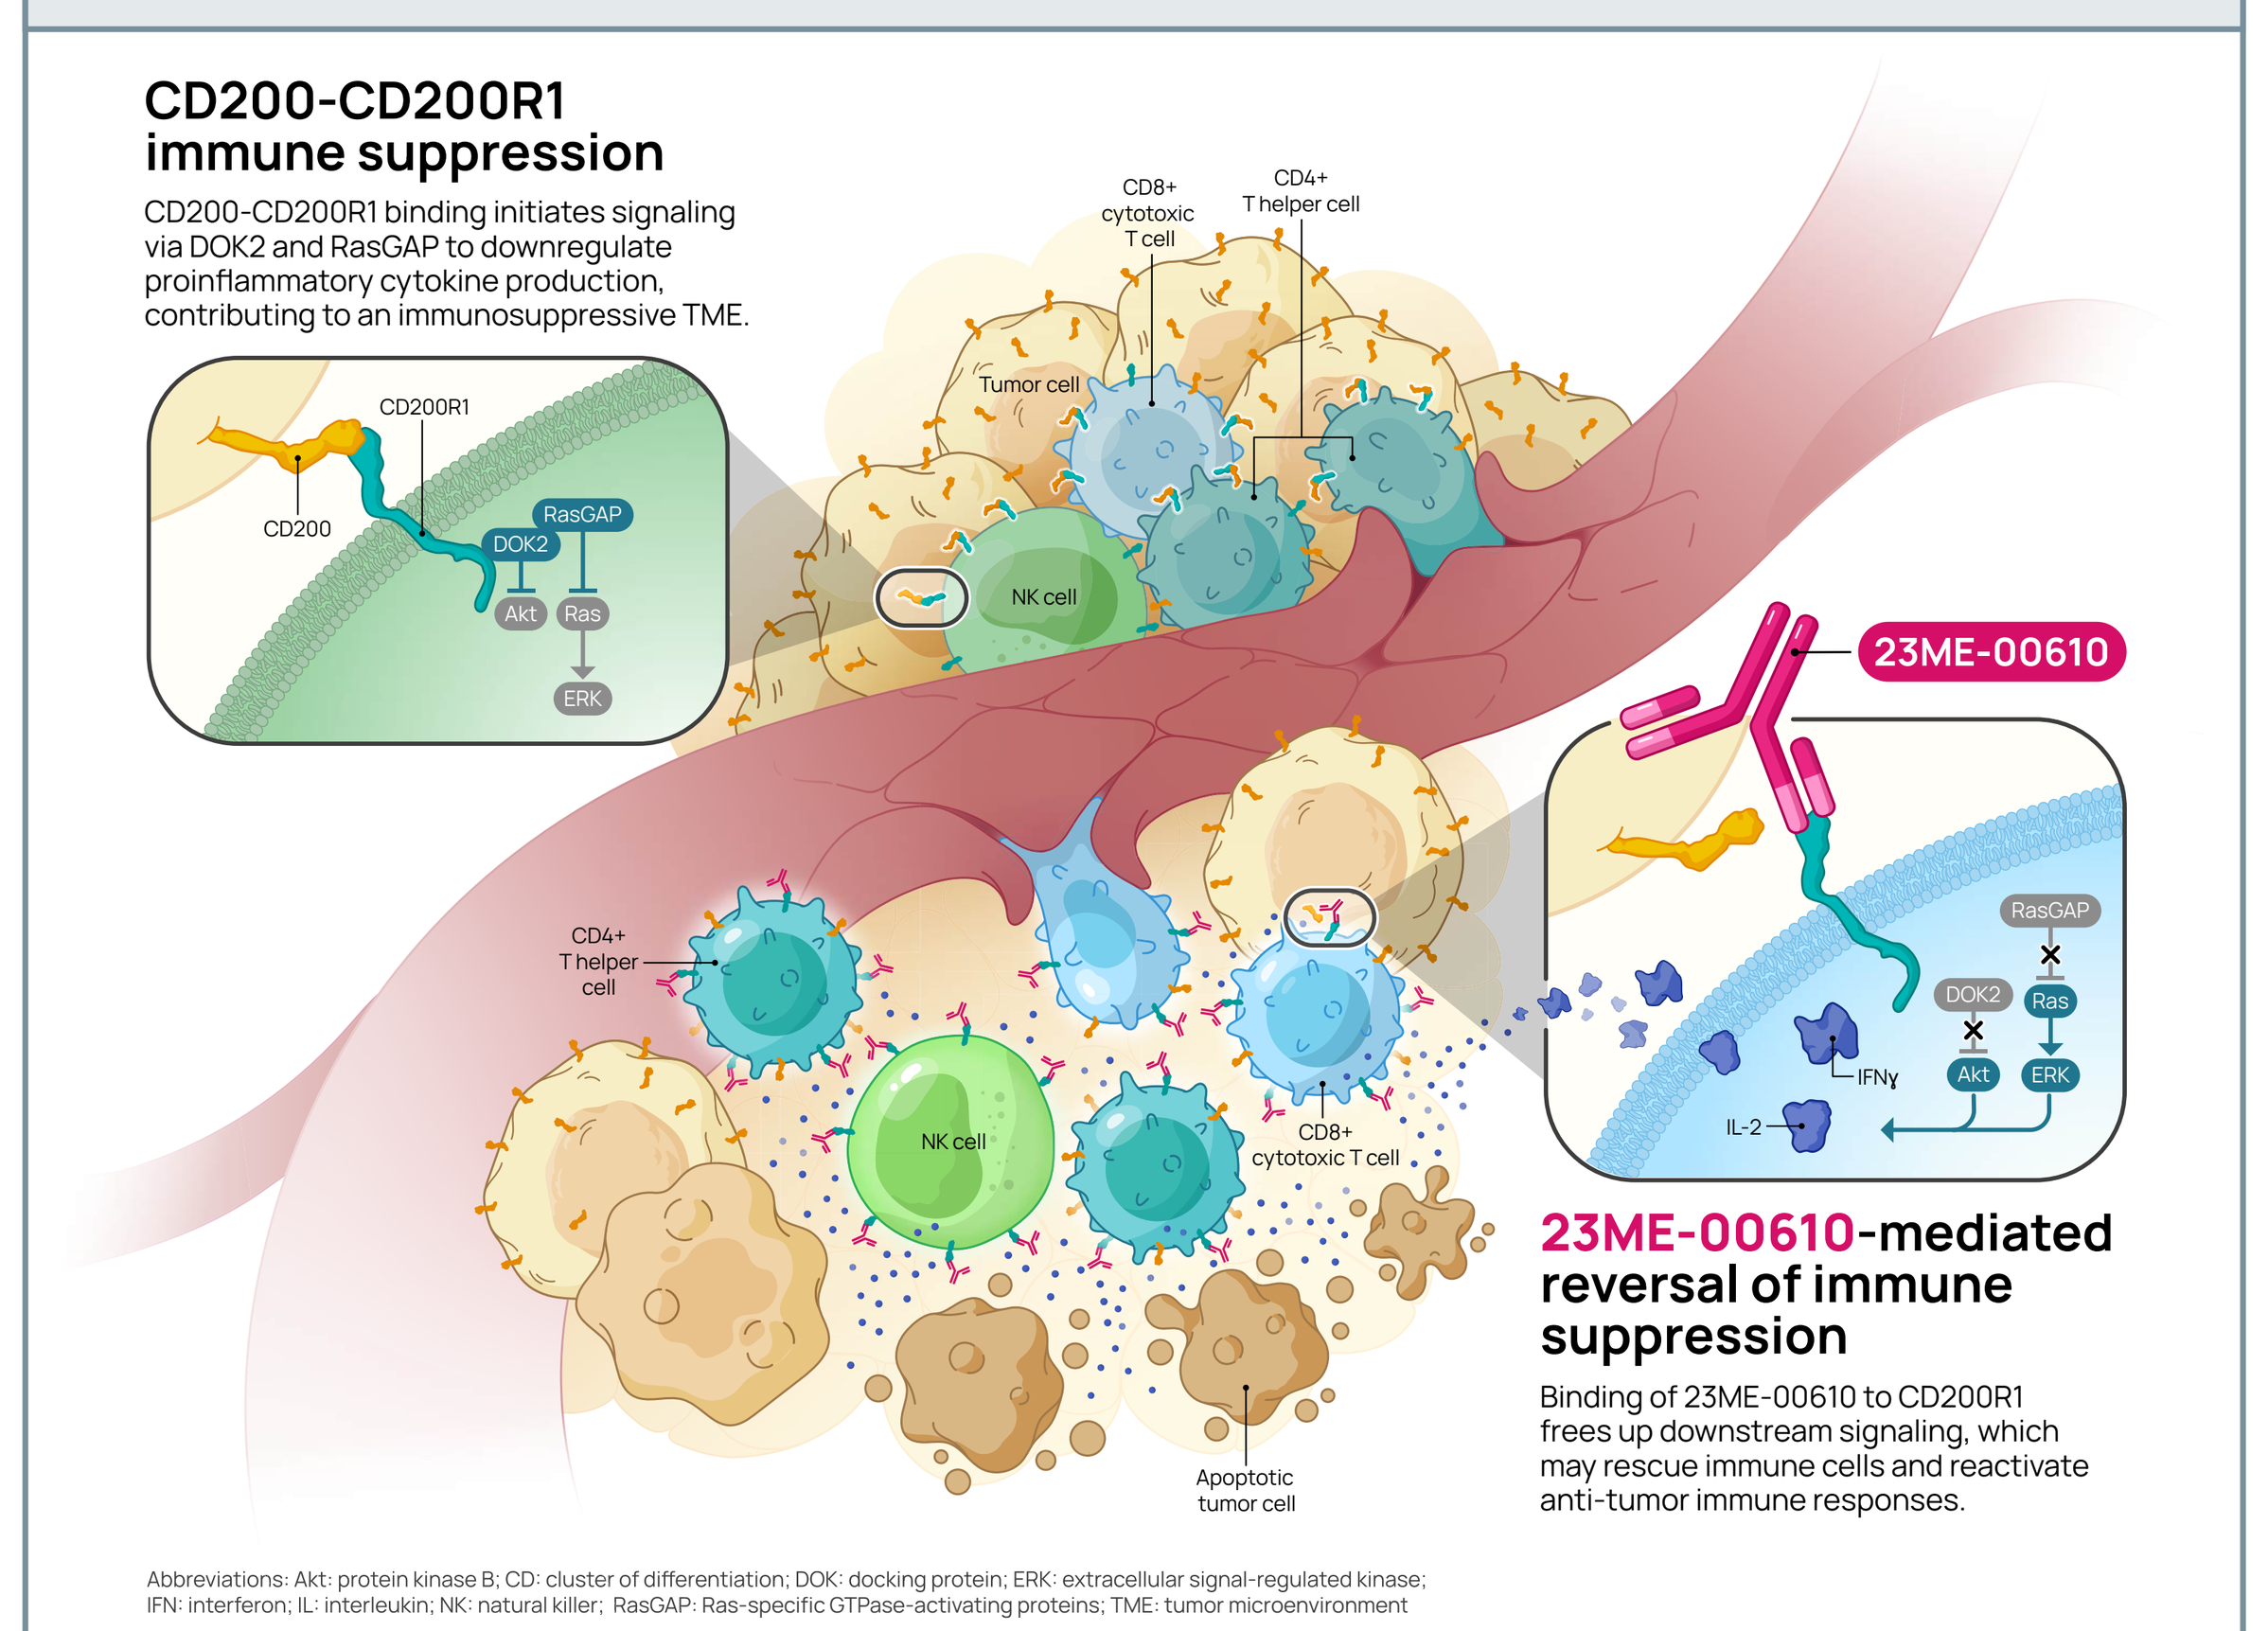
CD200-CD200R1 immune suppression
CD200-CD200R1 binding initiates signaling via DOK2 and RasGAP to downregulate proinflammatory cytokine production, contributing to an immunosuppressive TME.
CD200R1
CD200
RasGAP
DOK2
Akt
Ras
ERK
CD8+ cytotoxic T cell
CD4+ T helper cell
Tumor cell
NK cell
23ME-00610
IFNγ
IL-2
Apoptotic tumor cell
23ME-00610-mediated reversal of immune suppression
Binding of 23ME-00610 to CD200R1 frees up downstream signaling, which may rescue immune cells and reactivate anti-tumor immune responses.

Abbreviations: Akt: protein kinase B; CD: cluster of differentiation; DOK: docking protein; ERK: extracellular signal-regulated kinase; IFN: interferon; IL: interleukin; NK: natural killer; RasGAP: Ras-specific GTPase-activating proteins; TME: tumor microenvironment

RESULTS

Table 1. Baseline Characteristics and Patient Demographics

Characteristic	N = 16	Characteristic	N = 16
Age, median years (range)	56.5 (34-76)	Primary Site of Ovarian Cancer, n (%)	
Race, n (%)		Ovary	13 (81.3%)
White	15 (93.8%)	Peritoneum	2 (12.5%)
Native American/Alaska Native	1 (6.3%)	Mesonephric	1 (6.3%)
Ethnicity, n (%)		Histologic Subtypes, n (%)	
Not Hispanic or Latino	13 (81.3%)	High grade epithelial	7 (43.8%)
Unknown	3 (18.8%)	Clear cell	2 (12.5%)
Geographic Region, n (%)		Adenocarcinoma NOS	2 (12.5%)
USA	9 (56.3%)	Papillary serous	1 (6.3%)
Canada	7 (43.8%)	Mucinous	1 (6.3%)
ECOG [0 to 1], n (%)		Serous carcinoma NOS	1 (6.3%)
0	8 (50.0%)	Mixed histology (mucinous / endometroid)	1 (6.3%)
1	8 (50.0%)	Mesonephric adenocarcinoma	1 (6.3%)

Characteristic	N = 16
Months since Initial Diagnoses, median (range)	47 (11-167)
Prior Lines of Treatment, median (range)	4 (1-12)
Prior Systemic Cancer Therapy, n (%)	
Anti-PD-1/L1	3 (18.8%)
Anti-VEGF	12 (75.0%)
Platinum-based chemotherapy	16 (100.0%)
PARP inhibitor	9 (56.3%)
Other	11 (68.8%)

Figure 4. Overall 23ME-00610 AE profile, including TEAEs, TRAEs, and irAEs

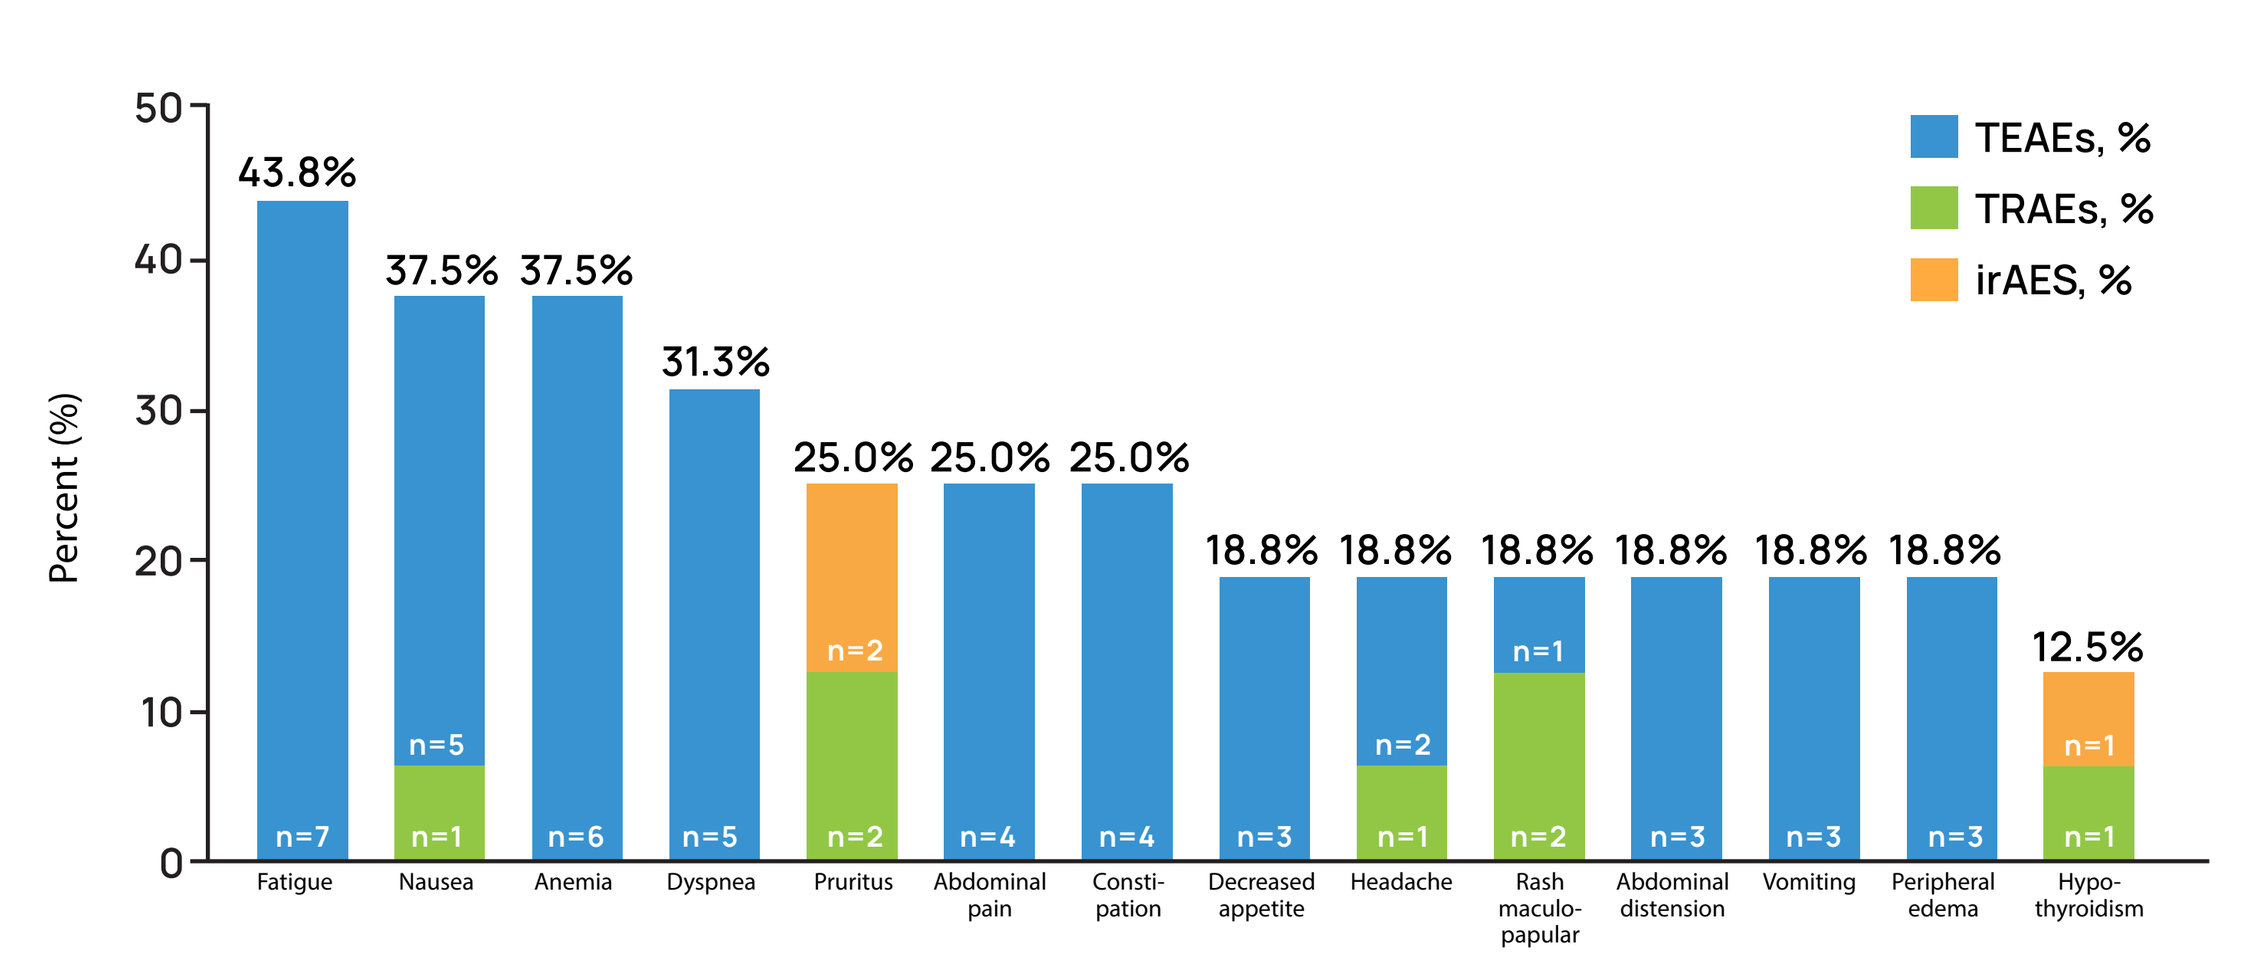
TEAEs, %
TRAEs, %
irAES, %
Percent (%)
43.8%
37.5%
37.5%
31.3%
25.0%
25.0%
25.0%
18.8%
18.8%
18.8%
18.8%
18.8%
18.8%
12.5%
Fatigue
Nausea
Anemia
Dyspnea
Pruritus
Abdominal pain
Constipation
Decreased appetite
Headache
Rash maculo-papular
Abdominal distension
Vomiting
Peripheral edema
Hypo-thyroidism

AEs observed in 2 or more patients (› 10%). Additional TEAEs of back pain, urinary tract infection, pleural effusion, aspartate amino-transferase increase, flank pain, and ascites were observed in N=2 patients each (12.5%).
TEAE, treatment emergent adverse event; TRAE, treatment related adverse event; irAE, immune related adverse event
Note: irAEs are considered related to study drug, and therefore not double counted in related category.

Figure 5. Tumor CD200 and CD200R1 Expression Shows ~50% Ovarian Patients with Moderate or High Tumor CD200

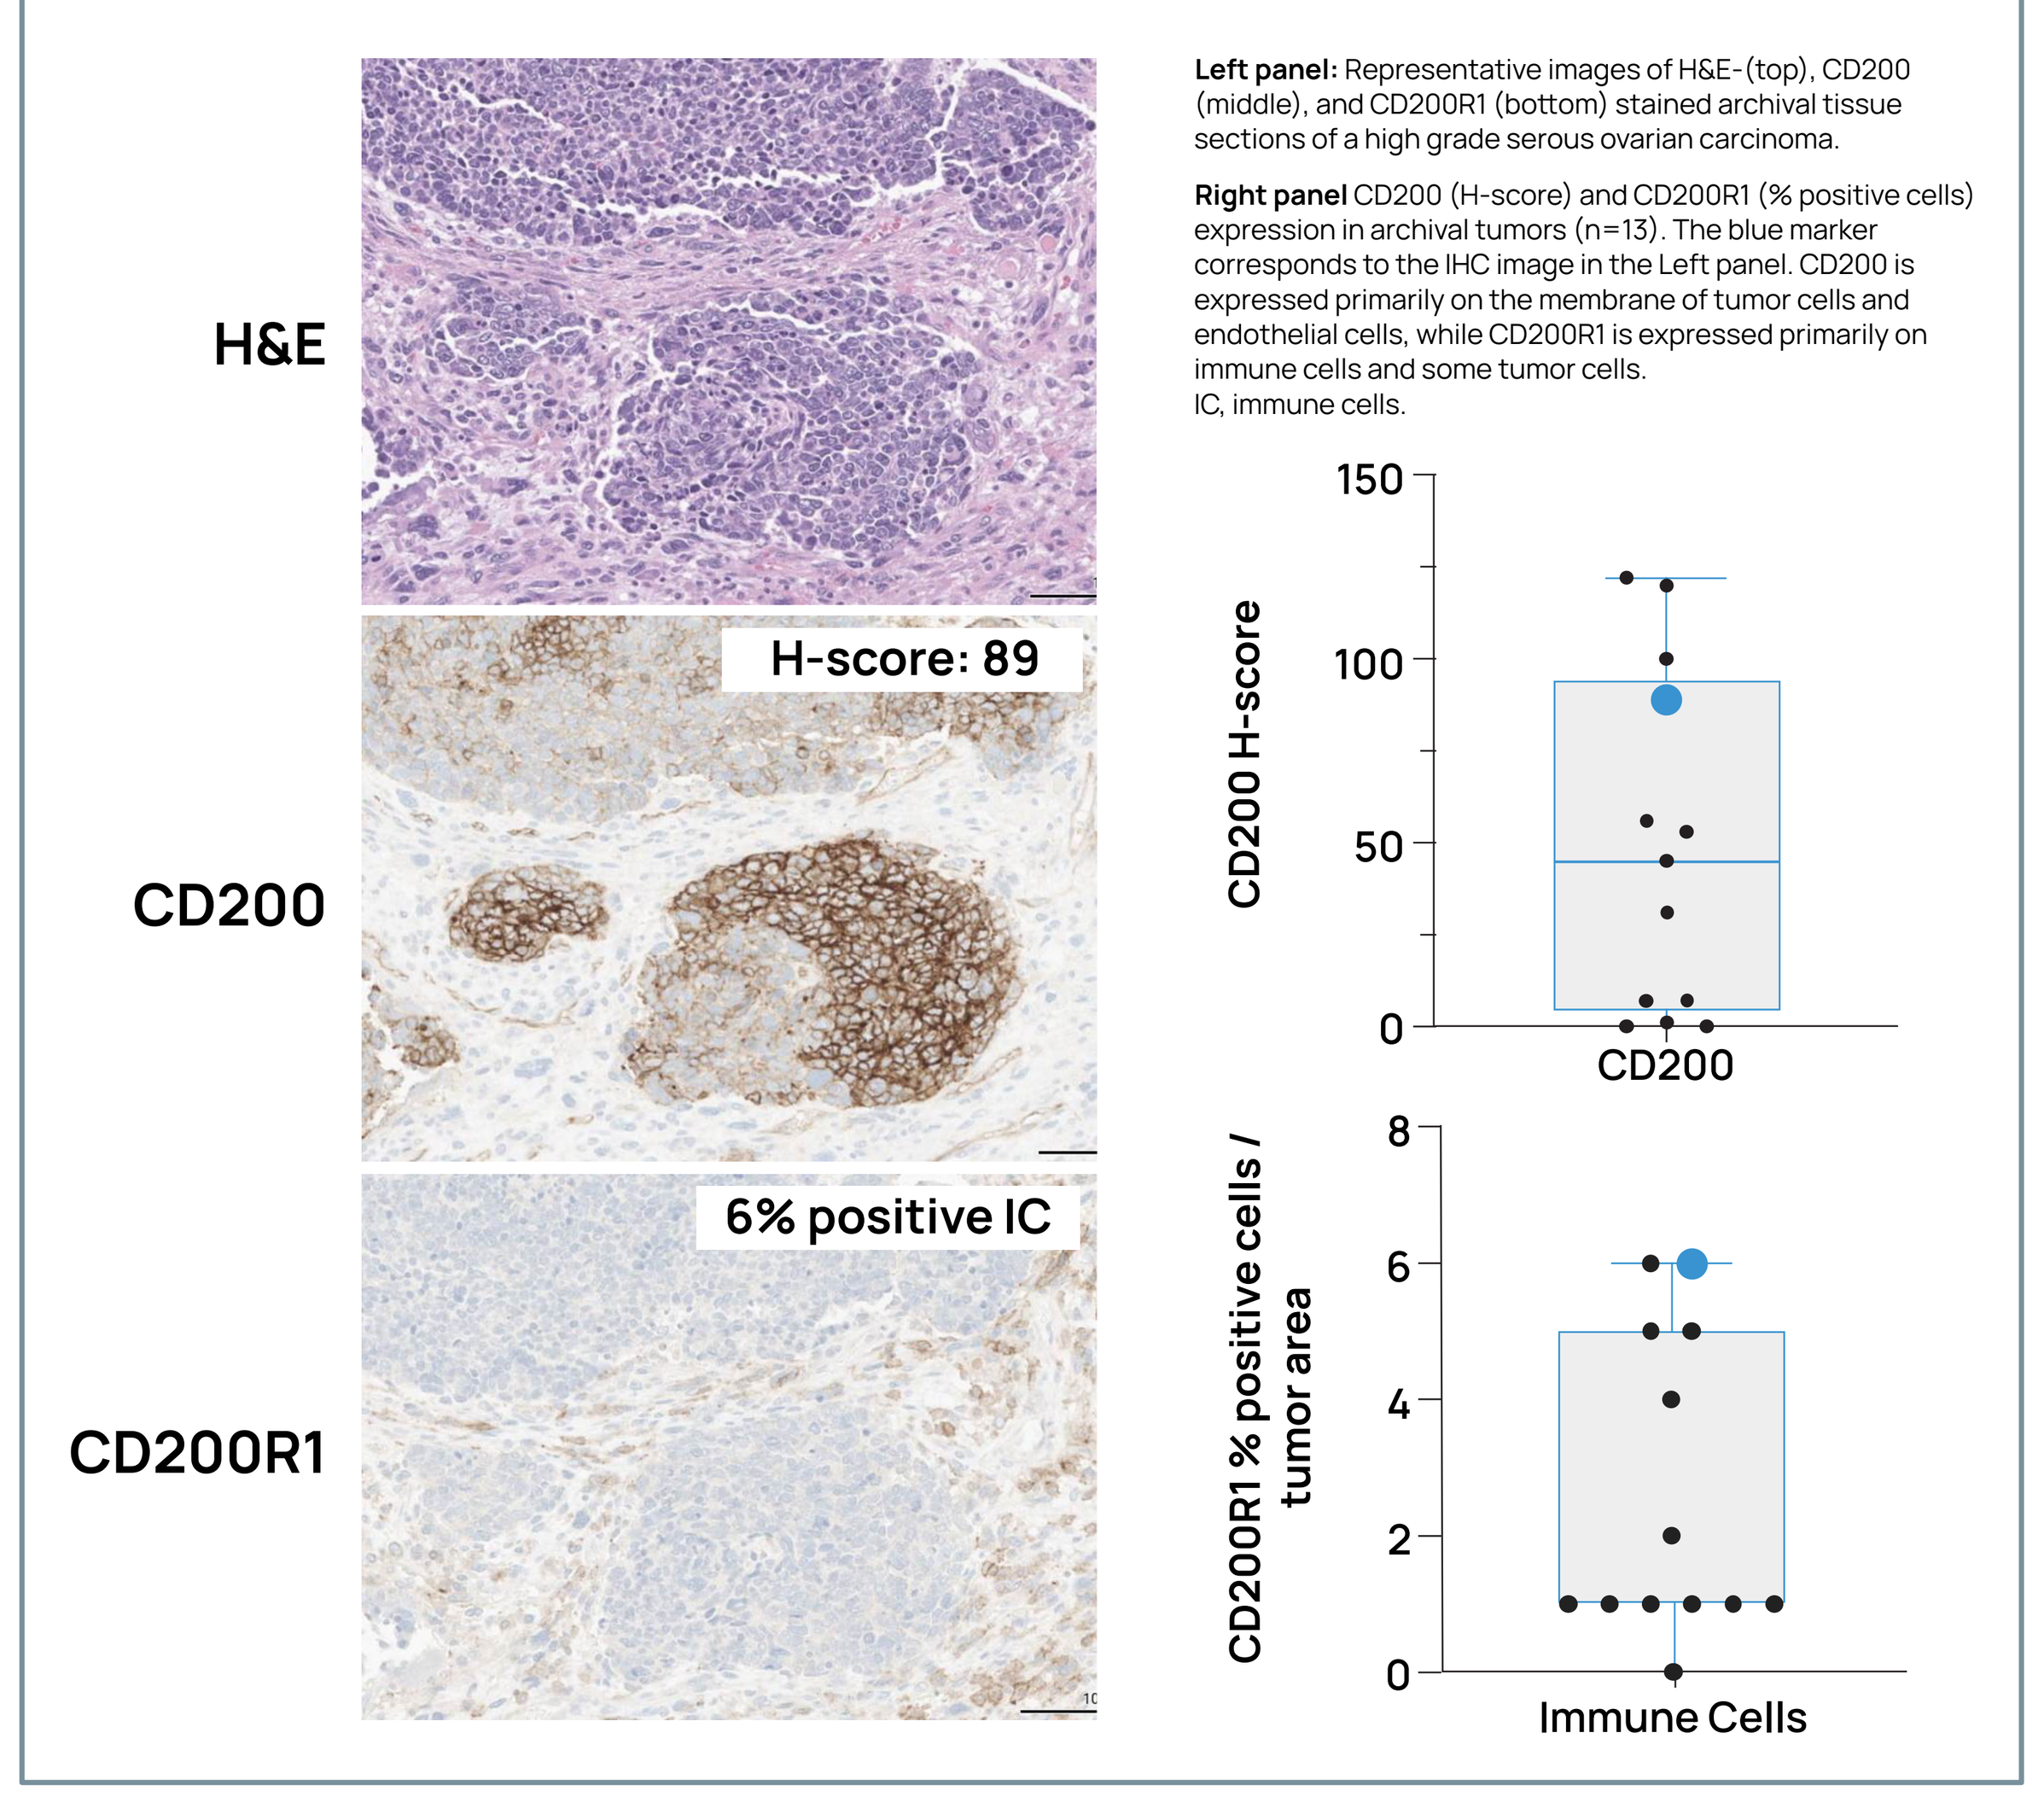
H&E
CD200
CD200R1
H-score: 89
6% positive IC
CD200 H-score
CD200
CD200R1 % positive cells / tumor area
Immune Cells

Left panel: Representative images of H&E-(top), CD200 (middle), and CD200R1 (bottom) stained archival tissue sections of a high grade serous ovarian carcinoma.

Right panel CD200 (H-score) and CD200R1 (% positive cells) expression in archival tumors (n=13). The blue marker corresponds to the IHC image in the Left panel. CD200 is expressed primarily on the membrane of tumor cells and endothelial cells, while CD200R1 is expressed primarily on immune cells and some tumor cells.
IC, immune cells.

Figure 6. 23ME-00610 Optimized PKPD at 1400 mg Q3W

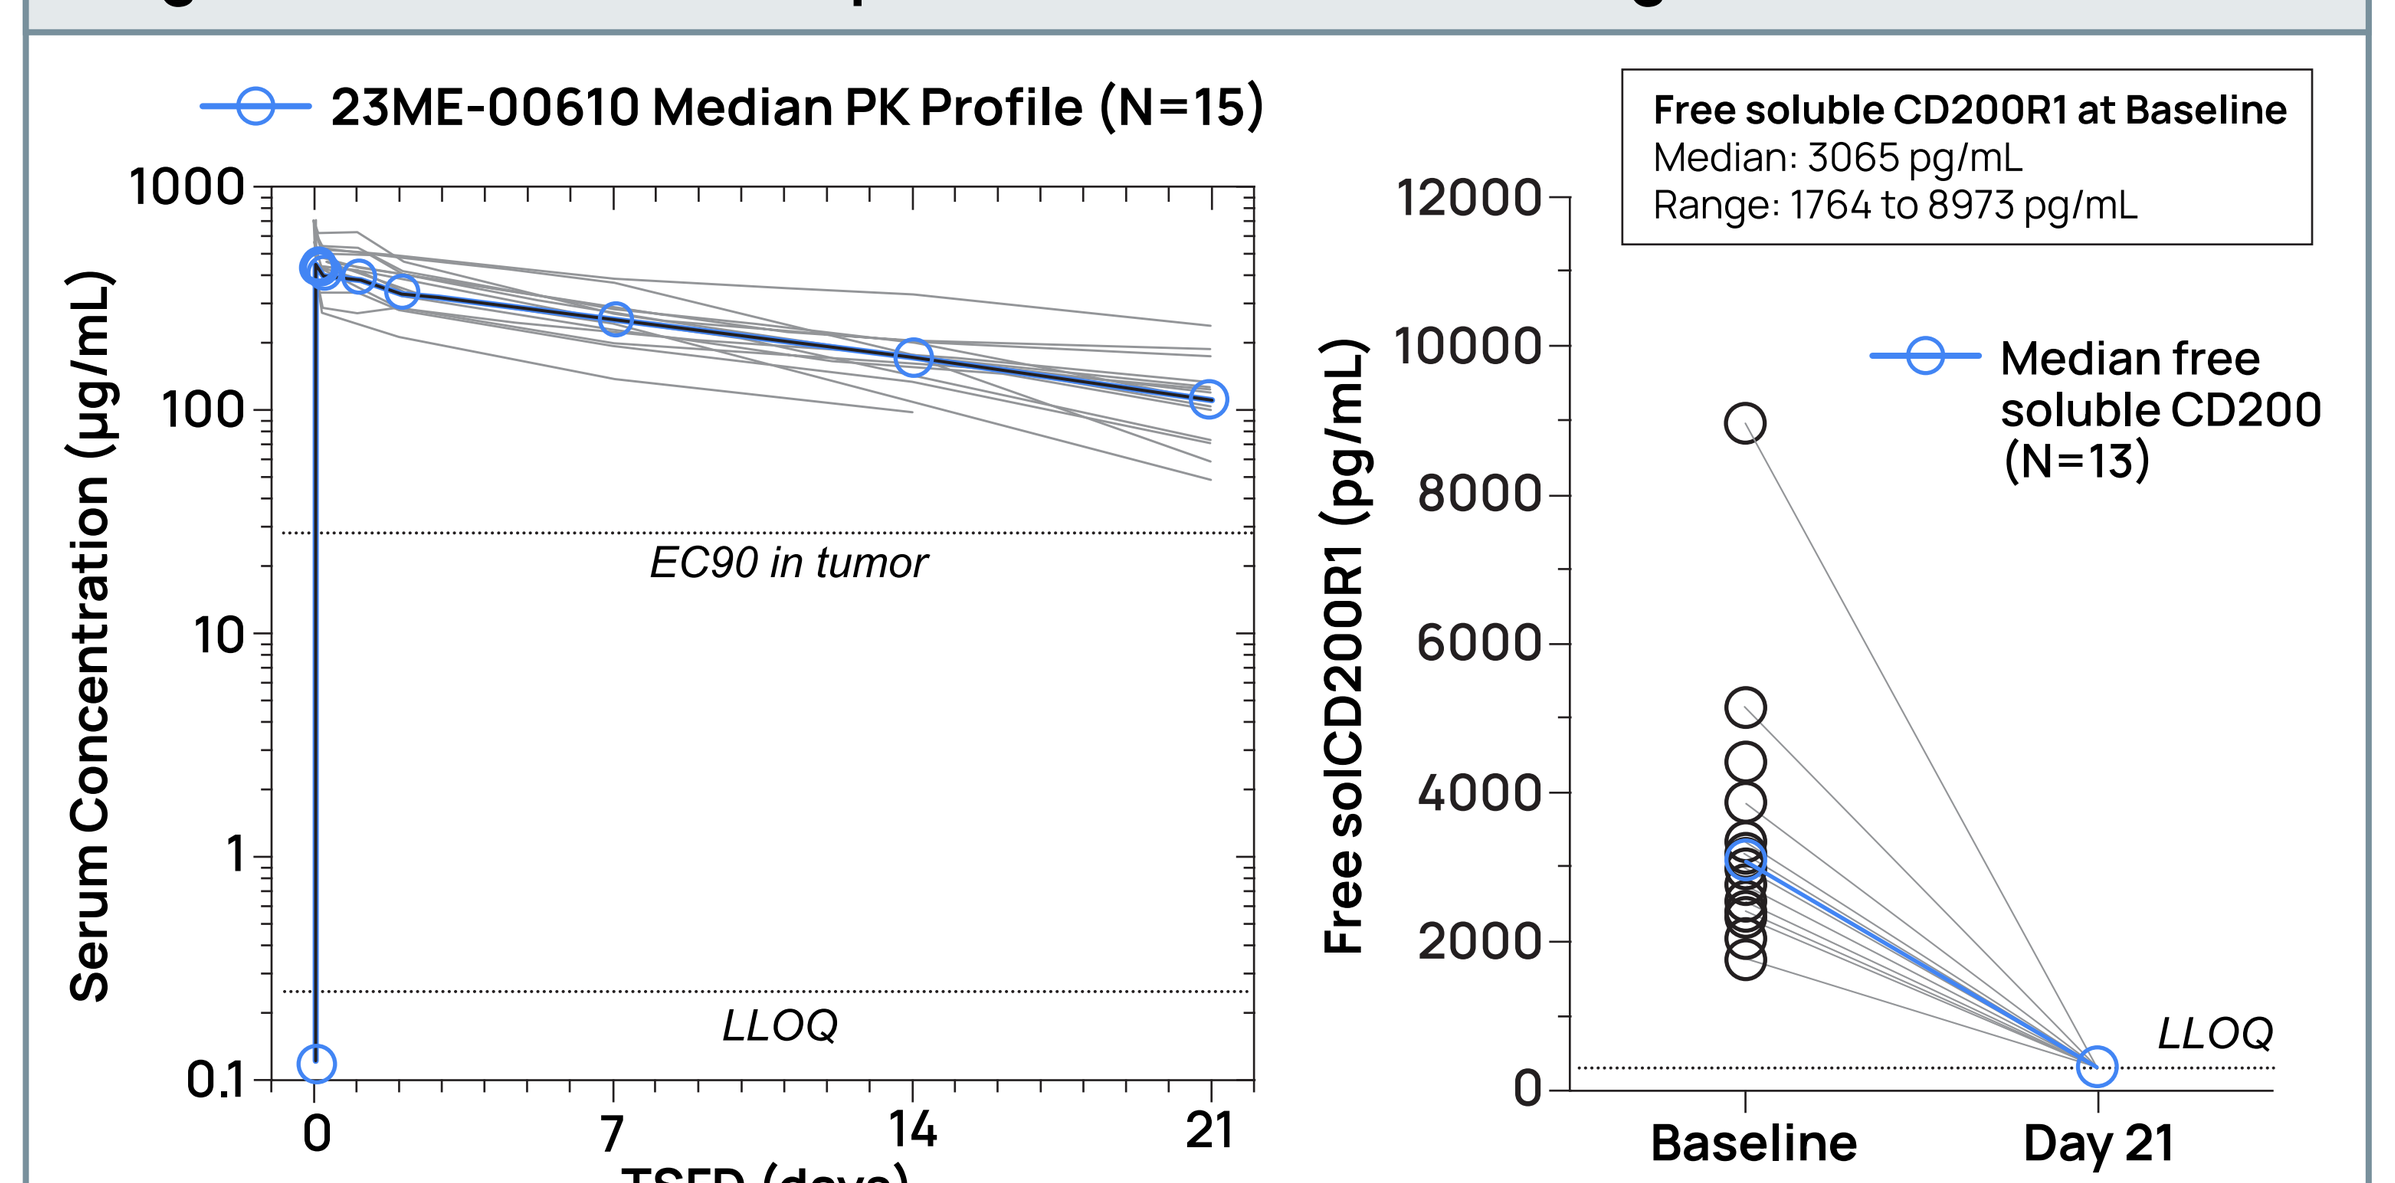
23ME-00610 Median PK Profile (N=15)
Serum Concentration (µg/mL)
EC90 in tumor
LLOQ
TSFD (days)
Free soluble CD200R1 at Baseline
Median: 3065 pg/mL
Range: 1764 to 8973 pg/mL
Median free soluble CD200 (N=13)
Free solCD200R1 (pg/mL)
Baseline
Day 21

PK data was unavailable for one patient at the time of analysis. Free soluble CD200R1 data were available for 13 patients due to Baseline and/or Day 21 data unavailable for 3 individuals. 23ME-00610 is administred IV every 3 week (Q3W); TSFD, time since first dose; Day 21 = C2D1 predose; Baseline was collected at C1D1 predose; LLOQ = lower limit of quantitation. BLQ values for PK concentration were set to 1/2 LLOQ. BLQ values for free soluble CD200R1 were set to LLOQ.

Figure 7. Best % Change of Summary of Diameters of Target Lesions

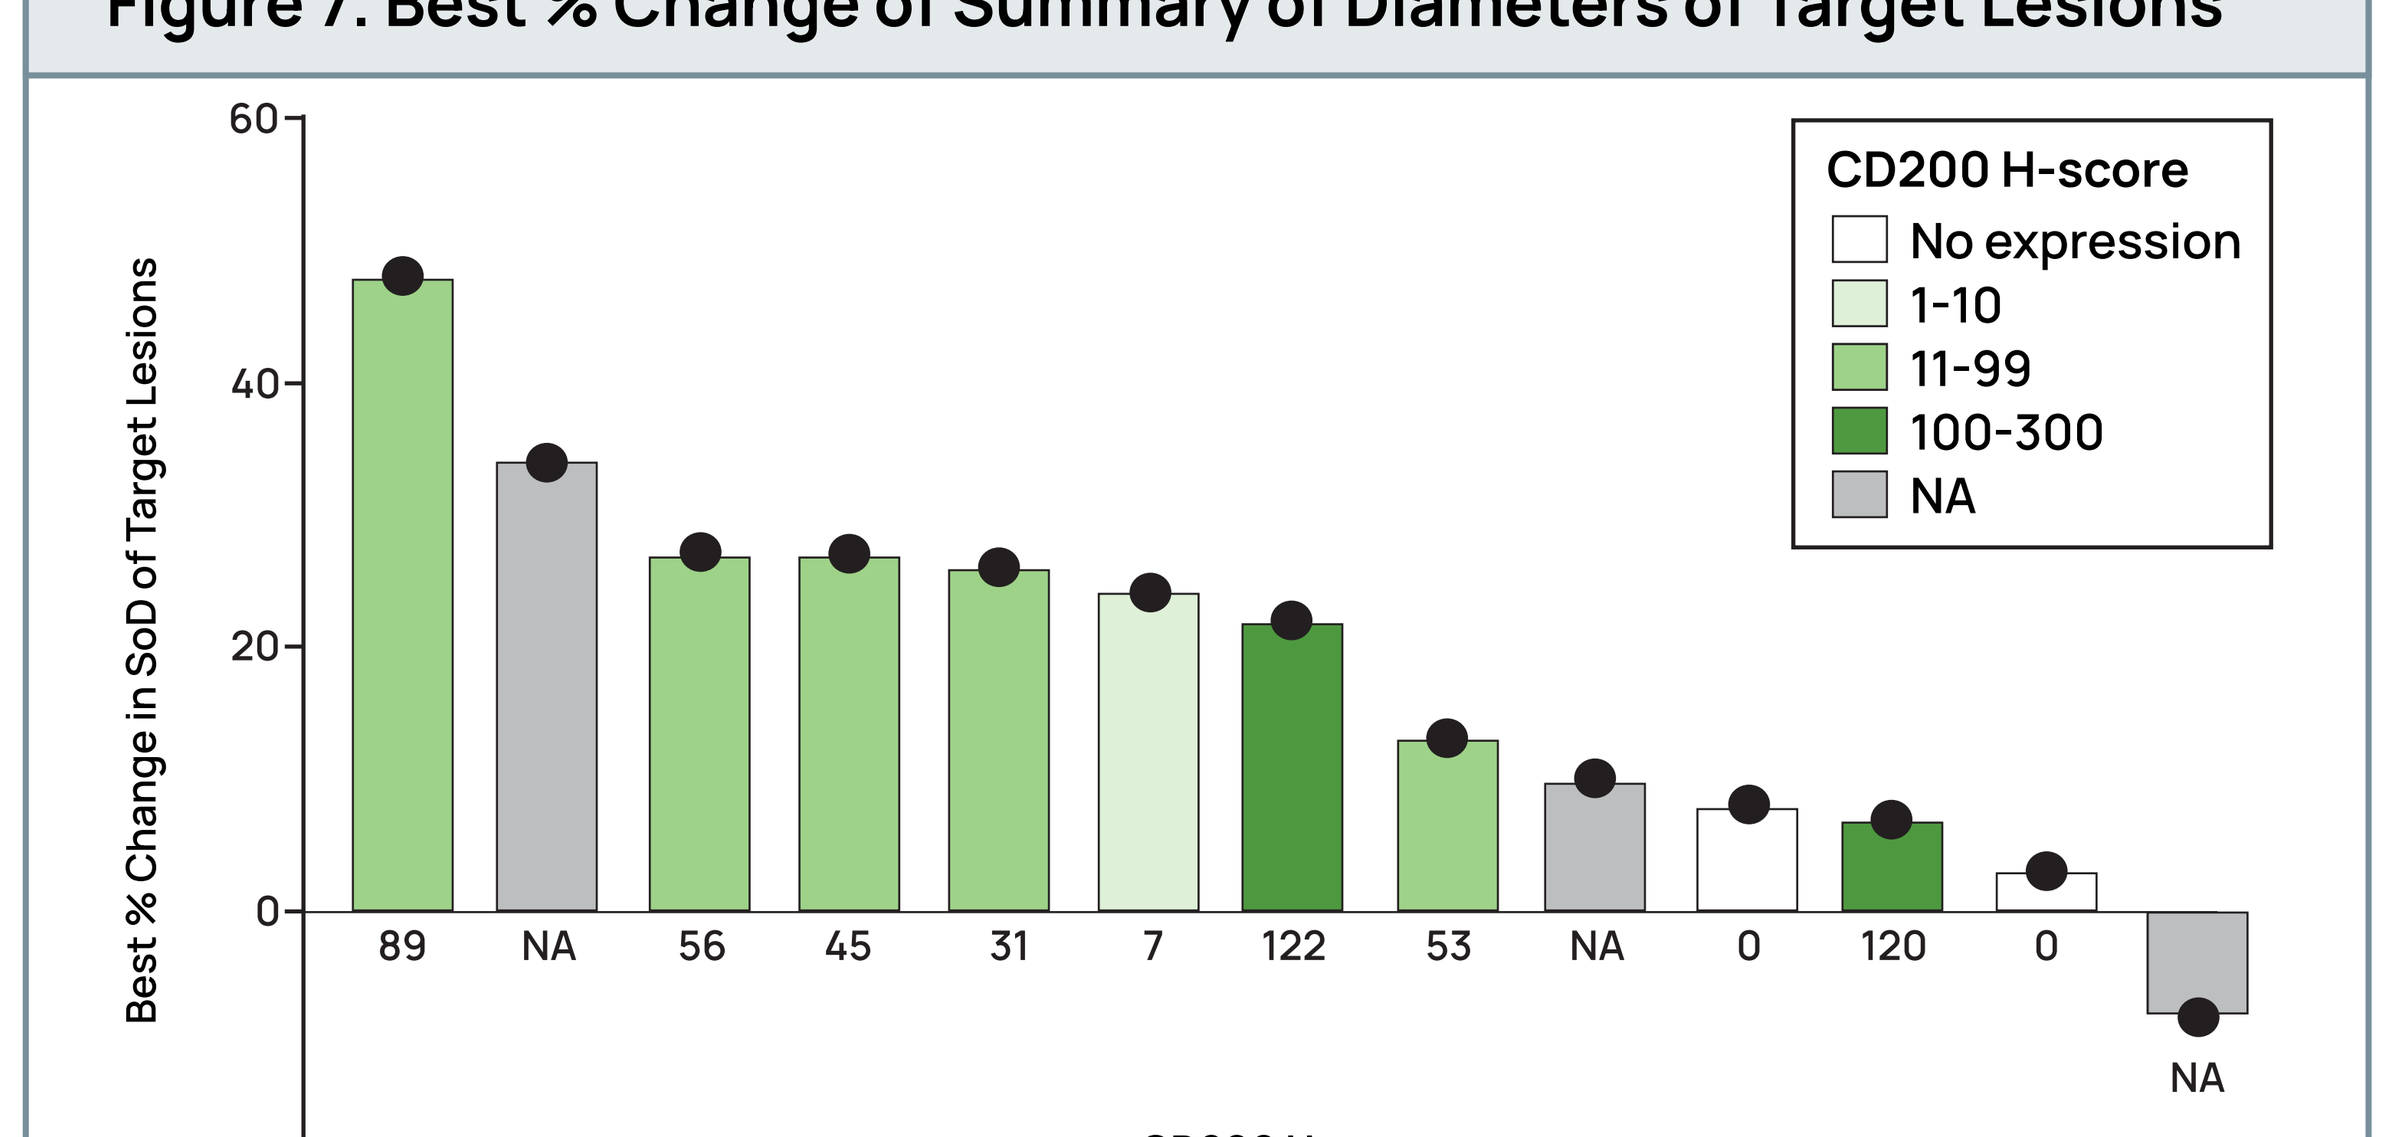
CD200 H-score
No expression
1-10
11-99
100-300
NA
Best % Change in SoD of Target Lesions
89
NA
56
45
31
7
122
53
NA
0
120
0
NA
CD200 H-score

Patients were sorted by Best % change in SoD of target lesions, and shaded based on their tumor CD200 H score. Patients had an average study drug exposure of 43 days (median 27 days; maximum 231 days). 3 subjects are not displayed on the plot due to missing post-baseline or not evaluable post-baseline SoD value.

METHODS

Figure 2. Phase 1/2a Study Design in Patients with Locally Advanced or Metastatic Solid Malignancies (NCT05199272)

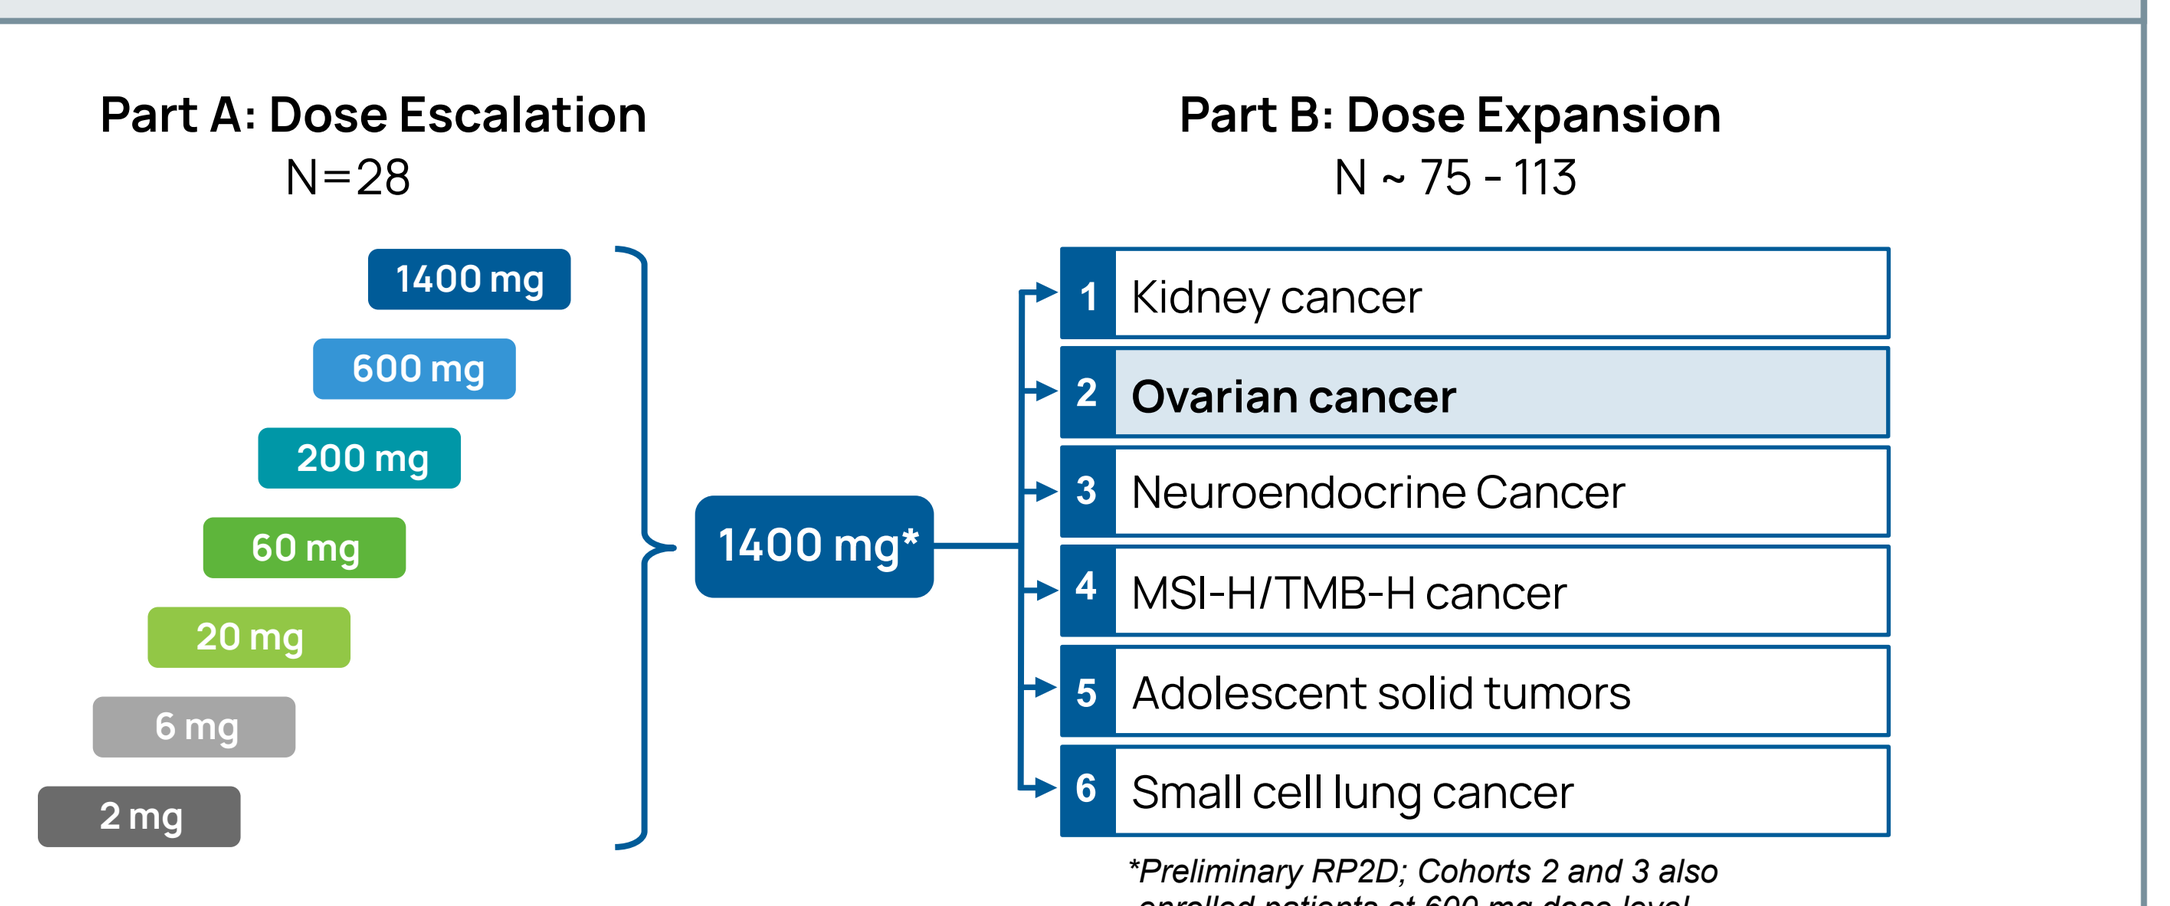
Part A: Dose Escalation
N=28
1400 mg
600 mg
200 mg
60 mg
20 mg
6 mg
2 mg
1400 mg*
Part B: Dose Expansion
N ~ 75 - 113
1 Kidney cancer
2 Ovarian cancer
3 Neuroendocrine Cancer
4 MSI-H/TMB-H cancer
5 Adolescent solid tumors
6 Small cell lung cancer
*Preliminary RP2D; Cohorts 2 and 3 also enrolled patients at 600 mg dose level

- 23ME-00610 administered IV every 21 days (Q3W)
- Expansions had target enrollment of 15 patients per cohort
- Ovarian cancer eligibility included patients with epithelial ovarian, fallopian or primary peritoneal carcinomas

Figure 3. Schedule of Assessments

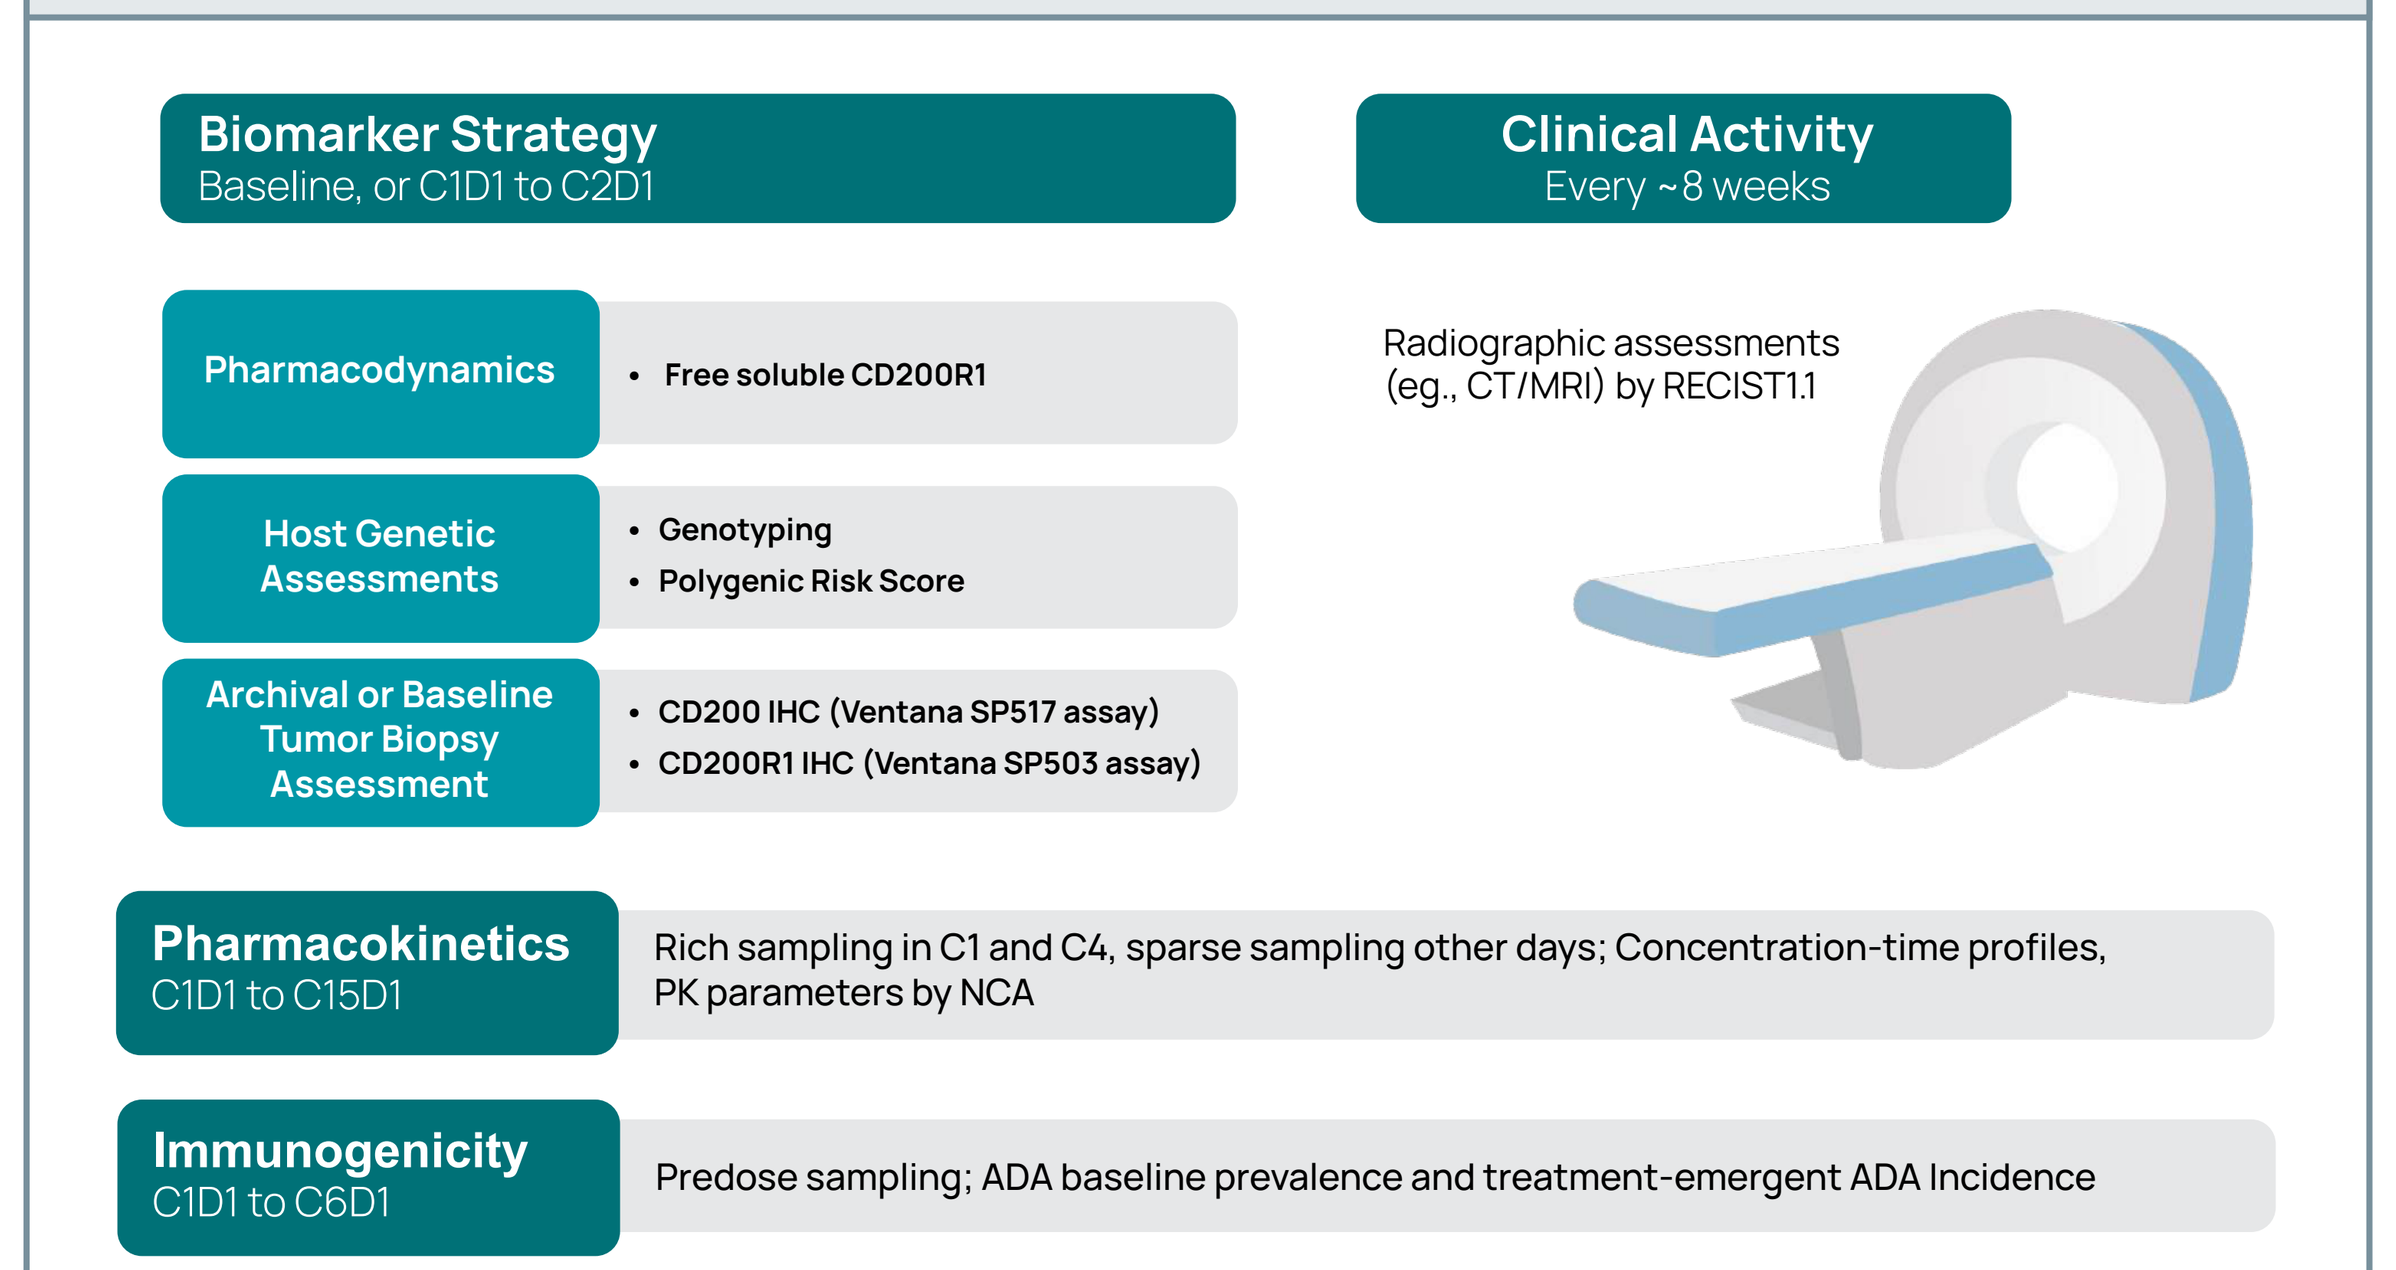
Biomarker Strategy
Baseline, or C1D1 to C2D1
Pharmacodynamics
Free soluble CD200R1
Host Genetic Assessments
Genotyping
Polygenic Risk Score
Archival or Baseline Tumor Biopsy Assessment
CD200 IHC (Ventana SP517 assay)
CD200R1 IHC (Ventana SP503 assay)
Clinical Activity
Every ~8 weeks
Radiographic assessments (eg., CT/MRI) by RECIST1.1
Pharmacokinetics
C1D1 to C15D1
Rich sampling in C1 and C4, sparse sampling other days; Concentration-time profiles, PK parameters by NCA
Immunogenicity
C1D1 to C6D1
Predose sampling; ADA baseline prevalence and treatment-emergent ADA Incidence

PATIENT VIGNETTE

Available Clinical Data Base Information

- 69-year old female with metastatic well-differentiated mesonephric adenocarcinoma (stage IA) initial diagnosis in 2017, surgically removed; patient has KRAS G12 mutation.
- Recurrence with tumor debulking in 2020 followed by doxorubicin for 5 months (best response stable disease) and trametinib for 20 months (best response with progressive disease).
- Patient enrolled on 23ME-00610-CLIN-001 with first dose in 2023 and has treatment ongoing for ›12 cycles as of April 1, 2024.

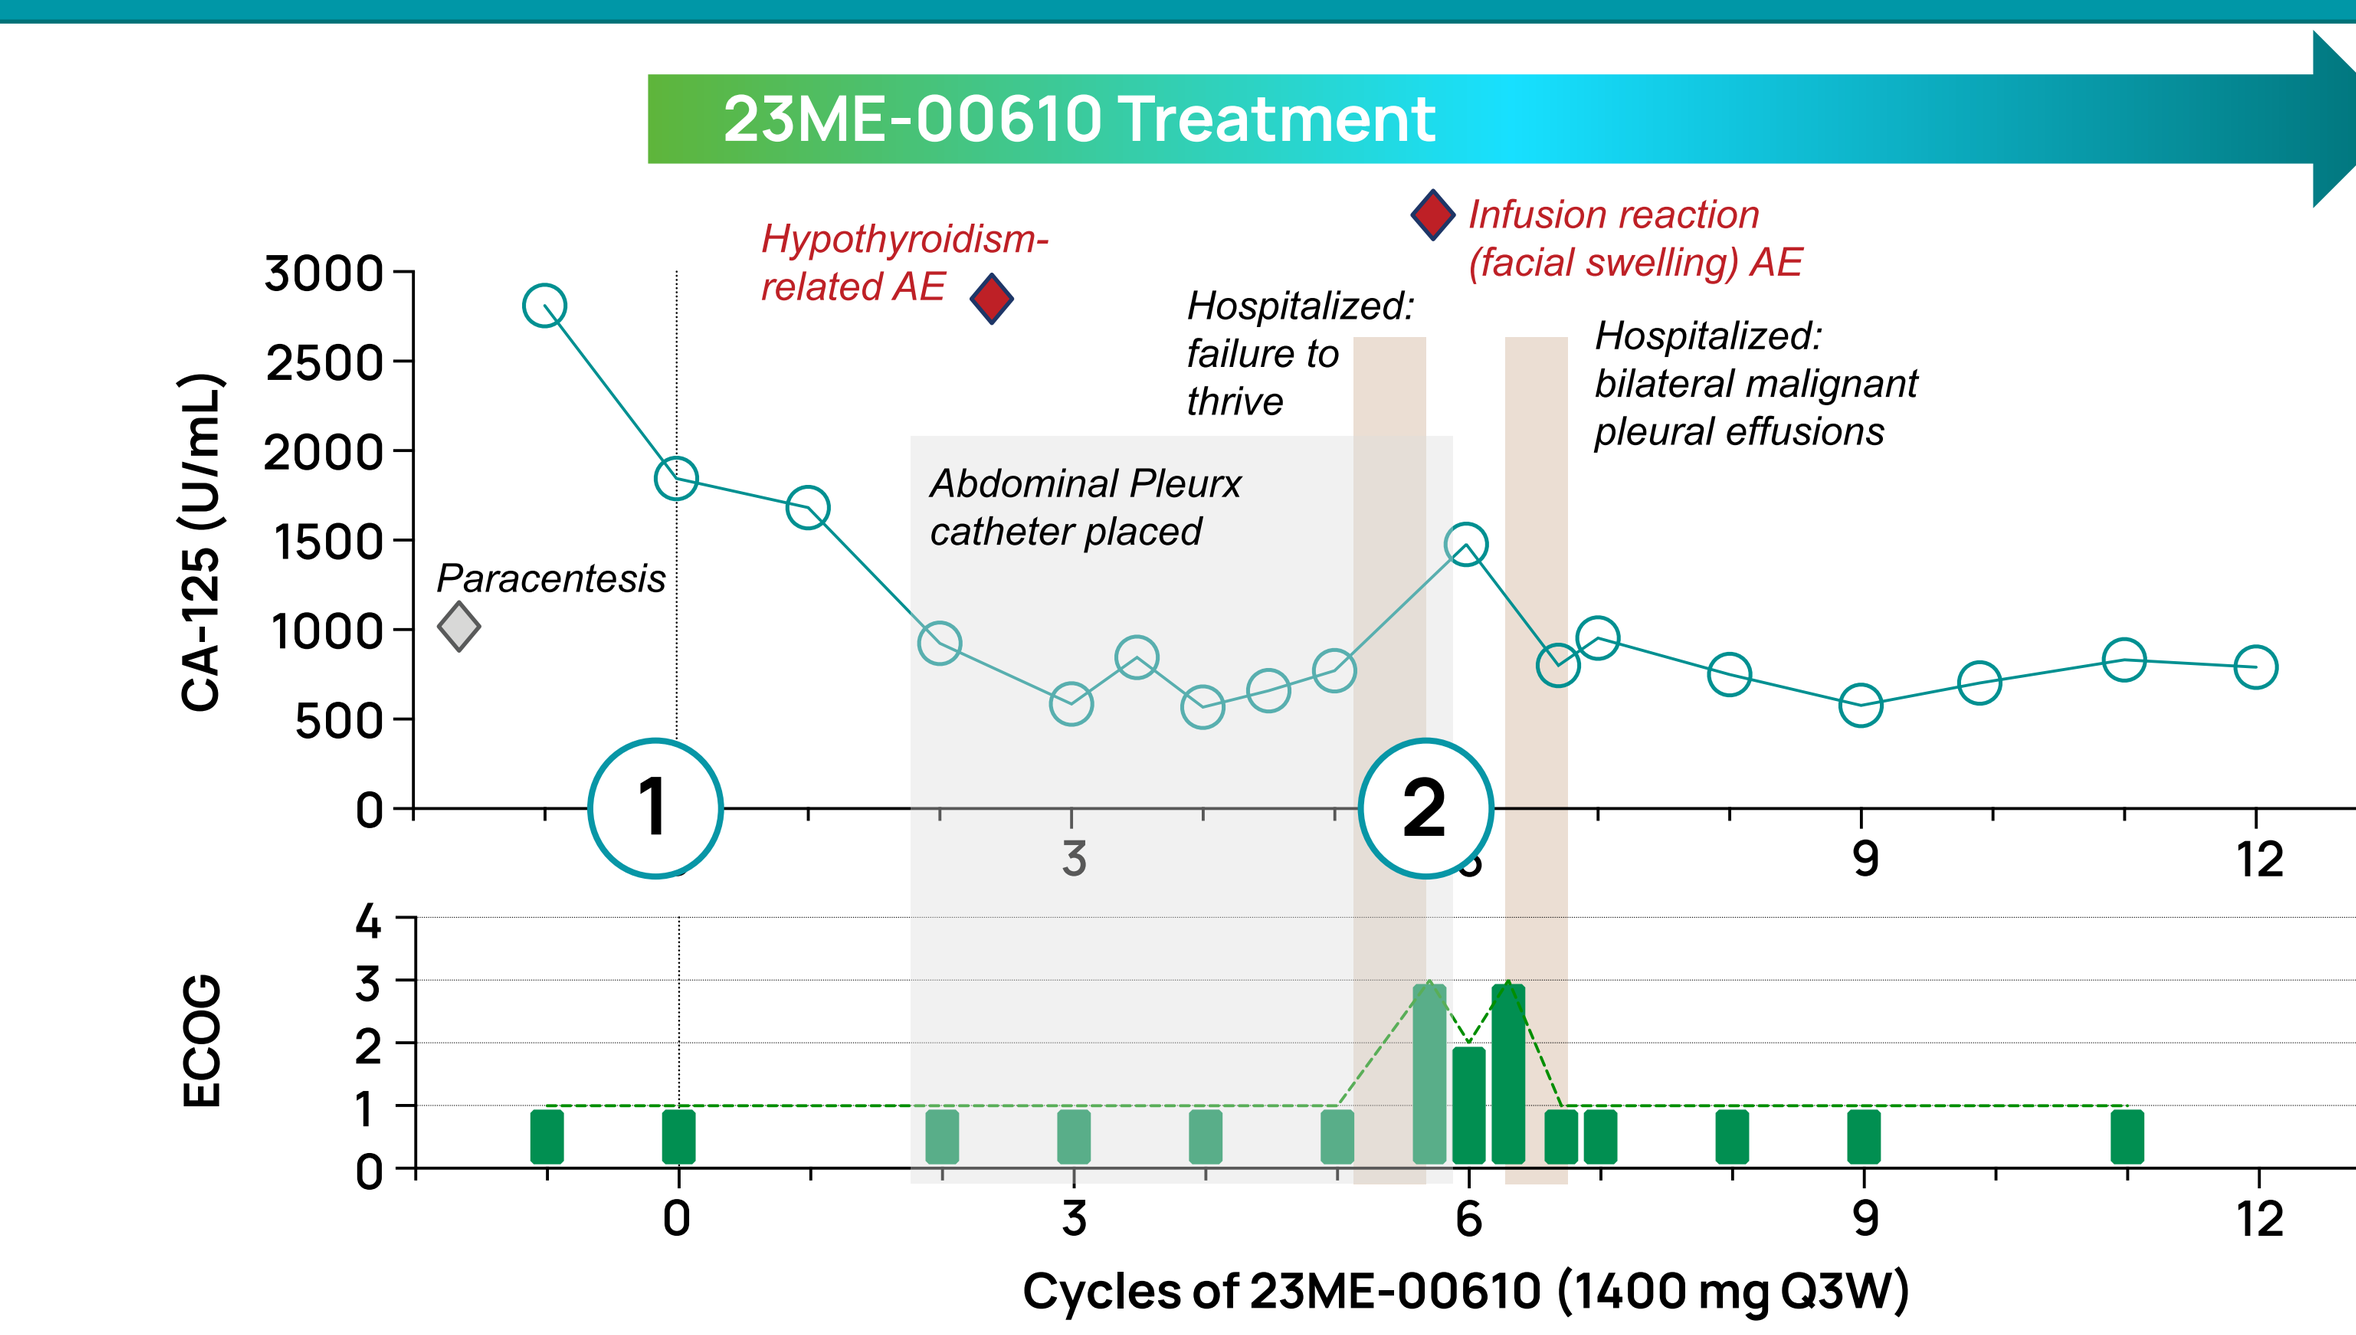
23ME-00610 Treatment
Hypothyroidism-related AE
Infusion reaction (facial swelling) AE
Hospitalized: failure to thrive
Hospitalized: bilateral malignant pleural effusions
Abdominal Pleurx catheter placed
Paracentesis
CA-125 (U/mL)
ECOG
Cycles of 23ME-00610 (1400 mg Q3W)

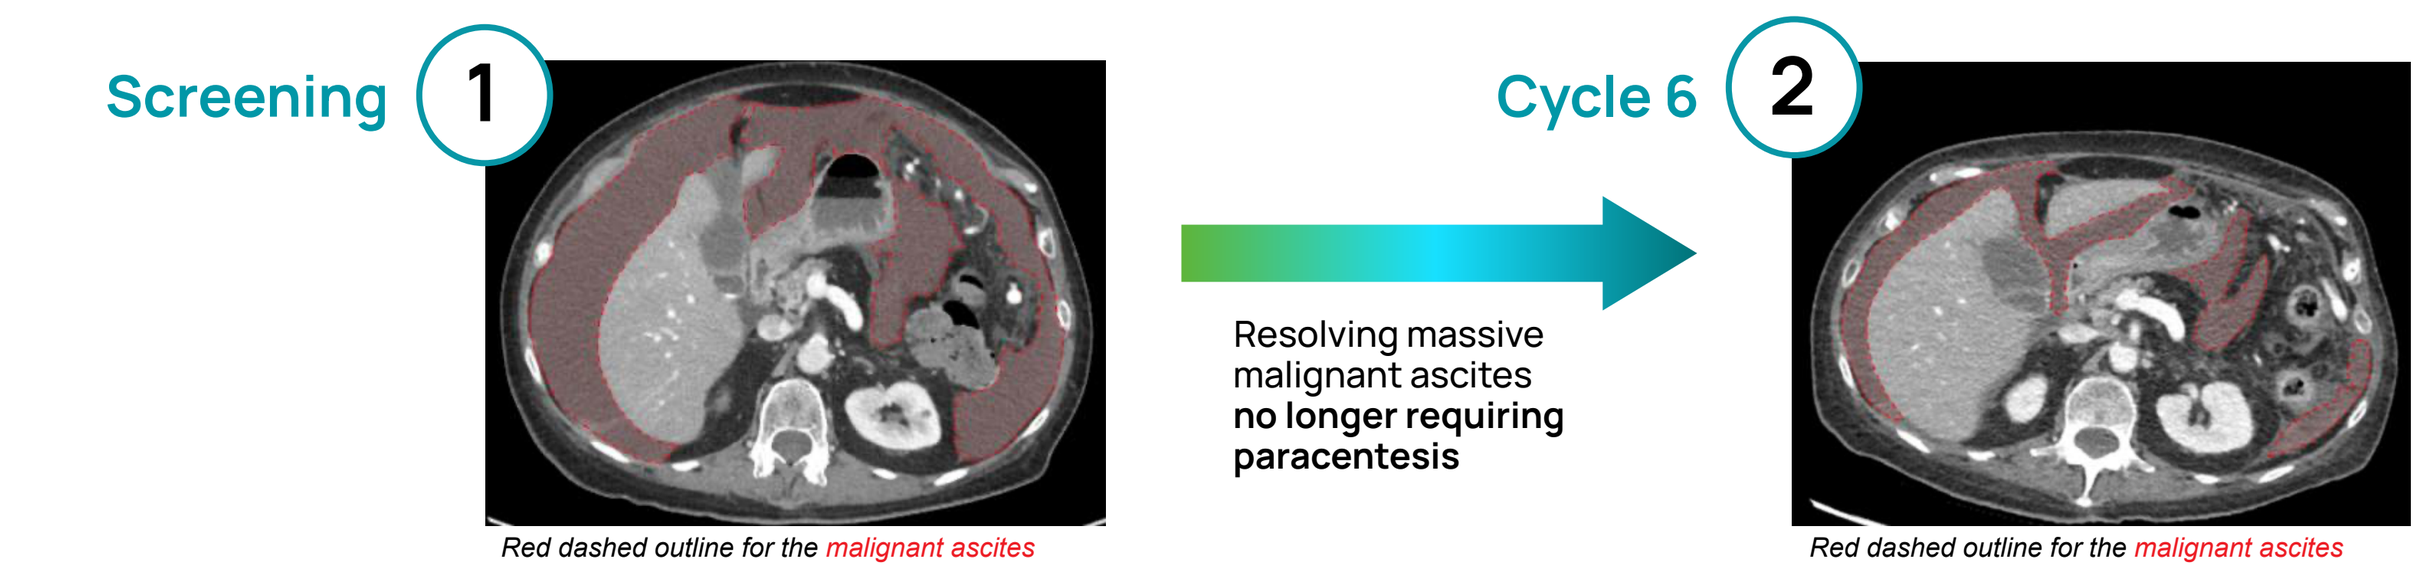
Screening
Cycle 6
Resolving massive malignant ascites no longer requiring paracentesis
Red dashed outline for the malignant ascites
Red dashed outline for the malignant ascites

Figure 8. Polygenic Risk Score (PRS) Values for Enrolled Trial Participants

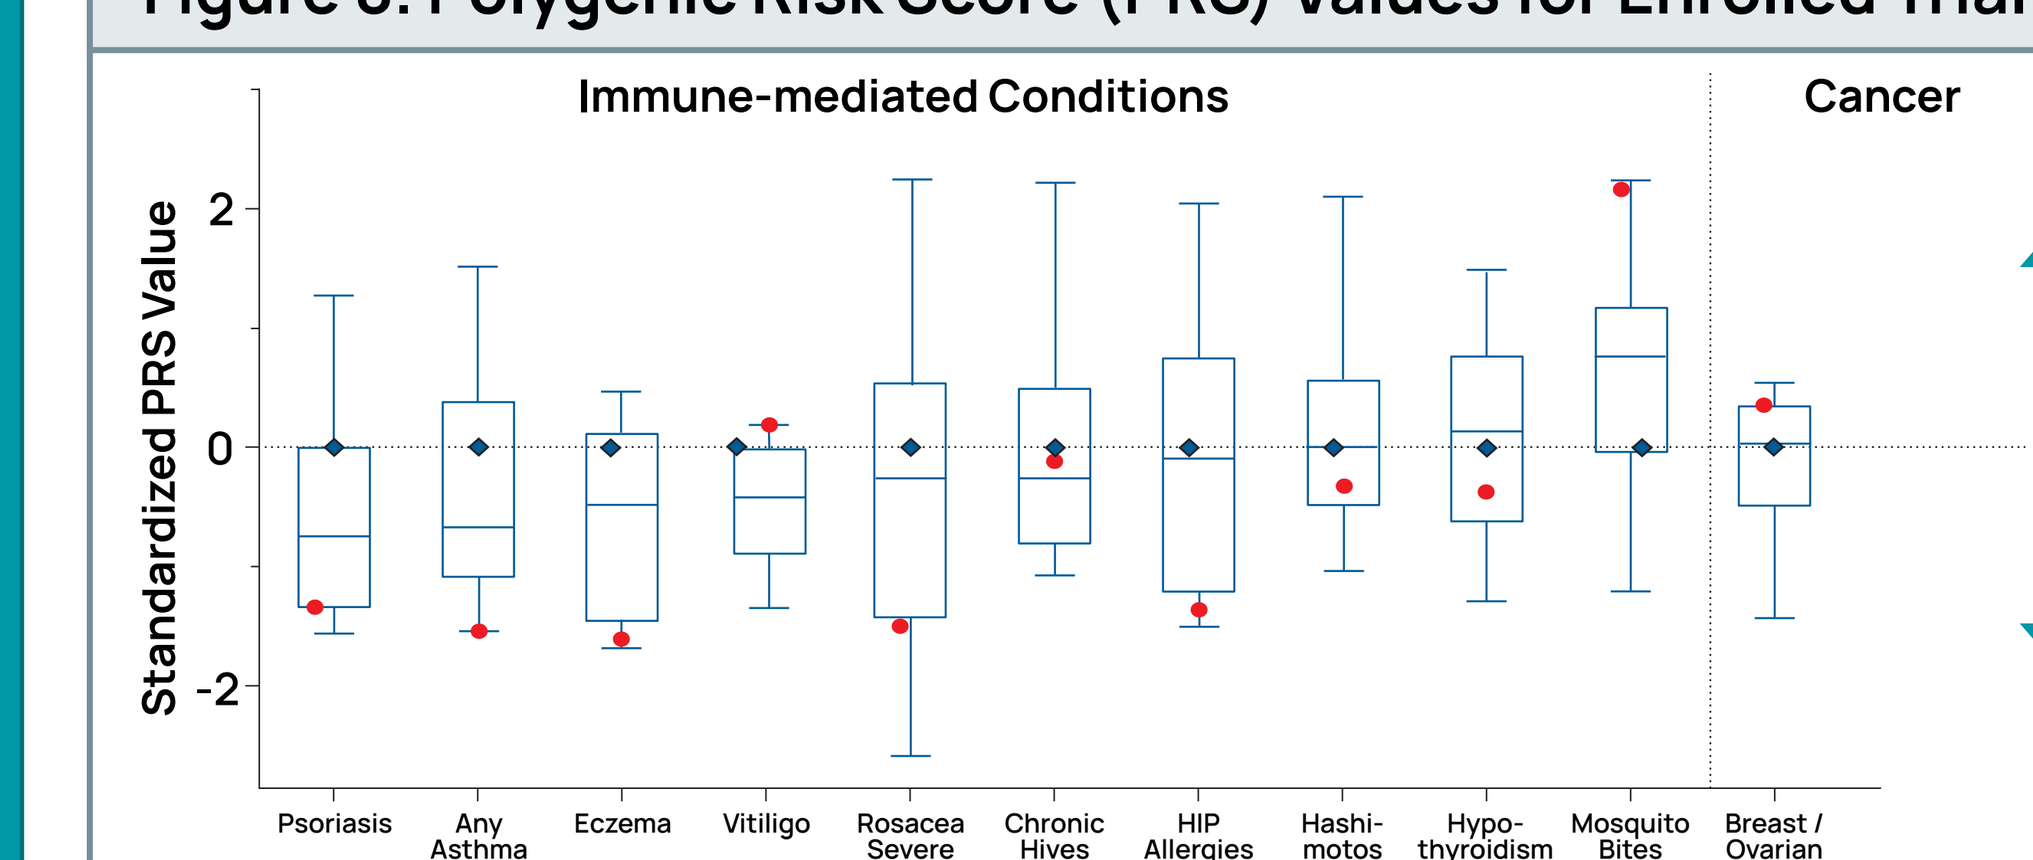
Immune-mediated Conditions
Cancer
Standardized PRS Value
Psoriasis
Any Asthma
Eczema
Vitiligo
Rosacea Severe
Chronic Hives
HIP Allergies
Hashimotos
Hypo-thyroidism
Mosquito Bites Itching
Breast / Ovarian Cancer
Higher risk
Lower risk

- This patient with mesonephric adenocarcinoma (red marker) has a lower genetic risk for some immune-mediated conditions relative to the reference population.
- PRS for immune and oncology relevant phenotypes for this patient generally trend in opposite directions. This "IO signature," opposing risk for cancer and immune conditions, is a genetic pattern for putative IO drug targets and led to the discovery of CD200R1 as a therapeutic target for cancer.

Standardized PRS values for genotyped patients of European ancestry (n = 10).
◆ = mean PRS for the 23andME European ancestry reference population,
● = patient vignette

CONCLUSIONS

- 23ME-00610 shows acceptable safety and tolerability and optimized PKPD at 1400 mg Q3W in ovarian cancer patients.
 - Related AEs were predominantly immune-related and included maculo-papular rash, pruritus, hypothyroidism, and nausea
 - irAEs were ≤ G2 in severity and generally dermatologic and thyroid in nature
 - No TRAEs ≥ G4 or AEs that led to death or discontinuation
 - Presumptive RP2D of 1400 mg achieves prespecified PK target and saturates solCD200R1, the PK profile generally supports Q3W dosing, and there was negligible ADA with no adverse impact on clinical activity
- Qualitative benefit and durable treatment duration (› 12 cycles) was observed for one patient with mesonephric adenocarcinoma.
 - 23ME-00610 treatment resulted in decreasing CA-125, substantial decrease of malignant ascites, and measurable tumor reduction (-8%).
 - Patient has a lower than typical risk for immune phenotypes (including psoriasis, asthma, eczema, and allergies) and high genetic risk for mosquito bite itching and breast or ovarian cancer

REFERENCES:

1. Fenaux J, et al. Oncoimmunology. 2023;12(1):2217737.
2. Mihrshahi R, et al. *J Immunol*. 2009;183(8):4879-4886.
3. Timmerman LM, et al. *PLoS One*. 2021;16(3):e0244770.
4. Misstear K, et al. *J Virol*. 2012;86(11):6246-6257.
5. Salek-Ardakani S, et al. *Eur J Immunol*. 2019;49(9):1380-1390.
6. Choueiry F, et al. *J Immunother Cancer*. 2020;8:e000189.
7. Moreaux J, et al. *Biochem Biophys Res Commun*. 2008;366:117-122.
8. Vathiotis IA, et al. *Cancers (Basel)*. 2021;13:1024.
9. Love JE et al. *Am J Clin Pathol*. 2017;148:236-242.
10. Kummar S, et al. *Cancer Res*. 2023;83(8_Supplement):CT174.
11. Rasco D, et al., *J for Immunother Cancer*. 2023;11:doi: 10.1136/jitc-2023-SITC2023.0619.
12. Glatt DM, et al., *J Immunother Cancer*. 2023;11:doi: 10.1136/jitc-2023-SITC2023.0609.